ECCMID 2018
Poster #P0157

Disk Content Assessment and Proposed Breakpoint Interpretive Criteria for Cefepime in Combination with AAI101

MD Huband[1], PR Rhomberg[1], KA Fedler[1], RK Flamm[1], P Knechtle,[2] S Shapiro[2]

[1] JMI Laboratories, Inc., North Liberty, Iowa, USA; [2] Allecra Therapeutics SAS, F-68300 Saint-Louis, France

Contact Information:
Michael D. Huband
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: michael-huband@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit https://www.jmilabs.com/data/posters/ECCMID2018-cefepime-disk.pdf

Charges may apply.
No personal information is stored.

Introduction

- AAI101 is a novel β-lactamase inhibitor (Figure 1) highly active against extended-spectrum β-lactamases (ESBLs), the primary mechanism of β-lactam resistance to oxyiminocephalosporins
- The combination of cefepime-AAI101 recently has completed Phase 2 clinical trials in complicated urinary tract infections
- Clinical and Laboratory Standards Institute (CLSI) M23 studies were conducted to select a cefepime-AAI101 disk content producing acceptable discrepancy rates against a collection of recent (mostly 2016) geographically diverse (mostly North America and Europe) clinical isolates and challenge strains of *Enterobacteriaceae*, including strains with β-lactamases characterized by whole genome sequencing

Materials and Methods

- A panel of 576 *Enterobacteriaceae* was assembled from clinical isolates obtained during worldwide surveillance studies predominantly in 2016 from North America and Europe
- Fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL, in combination with cefepime, were chosen for broth microdilution MIC determination based on prior microbiological and pharmacokinetic/pharmacodynamic studies
- For comparison purposes, cefepime breakpoint interpretive criteria were applied to cefepime-AAI101 combinations
- Four cefepime-AAI101 disk loads (30/30 µg, 30/20 µg, 30/15 µg, and 30/10 µg) initially were examined in duplicate against a challenge set of 58 *Enterobacteriaceae* clinical isolates, in parallel with cefepime (30 µg), meropenem (10 µg), and piperacillin-tazobactam (100/10 µg) controls in Tier 1 testing
- Two cefepime-AAI101 disk loads (30/20 µg and 30/10 µg) were chosen for Tier 2 testing
- Using the aforementioned two disk loads, recommended cefepime-AAI101 breakpoint interpretive criteria were generated according to CLSI guidelines M02-A12, M07-A10, M23-A4, and M100-S27
- Error-rate bounded analyses compared cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) MIC values to cefepime-AAI101 30/20 µg disk zone diameters
- Cefepime breakpoint interpretive criteria (CLSI and EUCAST) were applied to cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) MIC values for comparison and data analysis purposes

Figure 1 AAI101 compound structure

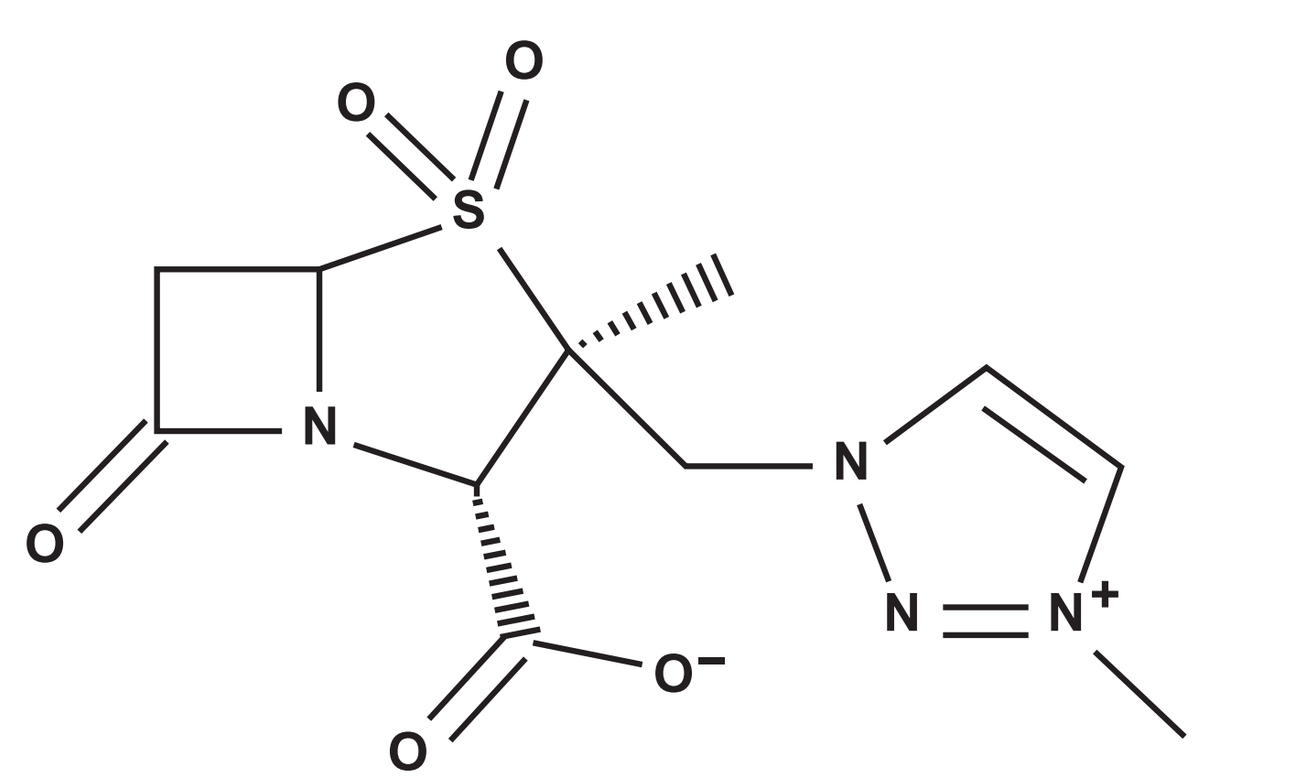

Figure 2 Proposed disk breakpoints based on the error-rate bounded method for cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) MIC *vs.* cefepime-AAI101 30/20 µg disks (CLSI cefepime MIC breakpoints for *Enterobacteriaceae* applied)

Cefepime-AAI101 (fixed 4 µg/mL)

		9	10	11	12	13	14	15	16	17	18	19	20	21	22	23	24	25	26	27	28	29	30	31	32	33	34	35	36	37	38	39	40
I+5	>128								1		1																						
I+4	128	2																															
I+3	64																																
I+2	32				2	1	1																										
I+1	16										1	3	2[a]																				
I	8														1		1																
I	4														2	1	3																
I-1	2															2[b]					1	1	1	1									
I-2	1												1[c]		1[c]				3	12	1	1	1	2		1	1						
I-3	0.5																	1	4	5	8	7	6	2	1	1	1						
I-4	0.25																		1	2	4	4	7	2	7	4	1		2				
I-5	0.12																1[d]	1			6	3	7	9	24	15	13	12	6	3			
I-6	0.06																						3	15	24	47	65	46	37	12	1	2	
I-7	0.03																			2	2		3	9	30	49	93	88	71	42	15	7	3
I-8	0.015																					2	1	9	3	5	21	31	27	14	11	6	2
I-9	0.008																							1	1		3	3		3	1		
I-10	≤0.004																																

Cefepime-AAI101 30/20 µg disks, zone diameter (mm)

[a] = *K. pneumoniae* (#942447; KPC-2, SHV-11, TEM-1); [b] = *K. pneumoniae* (#985823; OXA-48, SHV-28, CTX-M-15); [c] = *Serratia marcescens* (#937712); [d] = *E. coli* (#978205; KPC-3, TEM-1)
Calculated disk breakpoints of ≥25 mm (S) and ≤19 mm (R).

Figure 3 Proposed disk breakpoints based on the error-rate bounded method, of cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL) MIC *vs.* cefepime-AAI101 30/20 µg disks (CLSI cefepime MIC breakpoints for *Enterobacteriaceae* applied)

Cefepime-AAI101 (fixed 8 µg/mL)

		9	10	11	12	13	14	15	16	17	18	19	20	21	22	23	24	25	26	27	28	29	30	31	32	33	34	35	36	37	38	39	40
I+5	>128																																
I+4	128	2							1		1																						
I+3	64																																
I+2	32				2	1	1																										
I+1	16										1	3																					
I	8												2		1		1																
I	4															1	1																
I-1	2															2[a]	2[b]				1	1											
I-2	1												1[c]		3[c,d]				1	5	2	1	2	1		1	1						
I-3	0.5																	1	4	12	5	9	6	3	2	1	1						
I-4	0.25																		2	1	6	1	6	5	6	1							
I-5	0.12																		1	1	4	2	5	8	16	18	10	8	8	3			
I-6	0.06																1[e]	1			2	2	7	15	33	52	77	53	40	10	4	3	
I-7	0.03																			2	2		2	12	26	42	82	83	71	49	11	7	3
I-8	0.015																					2	1	6	7	7	26	35	24	12	13	5	2
I-9	0.008																										1	1					
I-10	≤0.004																																

Cefepime-AAI101 30/20 µg disks, zone diameter (mm)

[a] = *K. pneumoniae* (#985823; OXA-48, SHV-28, CTX-M-15); [b] = *Enterobacter cloacae* species complex (#952344; ACT-17); [c] = *S. marcescens* (#937712);
[d] = *K. pneumoniae* (#942544; KPC-2, SHV-28, CTX-M-15, OXA-1/30); [e] = *E. coli* (#978205; KPC-3, TEM-1)
Calculated disk breakpoints of ≥25 mm (S) and ≤19 mm (R)

Table 1 Cumulative percent inhibition results for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and comparators against a challenge set of 58 *Enterobacteriaceae* isolates (Tier 1)

Organism group (no. tested)/ antimicrobials	No. of isolates at MIC (µg/mL; cumulative %) 0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	>[a]	MIC_{50}	MIC_{90}
Enterobacteriaceae (58)[b]																
AAI101													0 0.0	**58** **100.0**	>128	>128
Cefepime-AAI101 (fixed 4 µg/mL)	8 15.5	8 29.3	8 43.1	5 51.7	3 56.9	5 65.5	2 69.0	3 74.1	7 86.2	**5** **94.8**	2 98.3	0 98.3	0 98.3	1 100.0	0.25	16
Cefepime-AAI101 (fixed 8 µg/mL)	7 13.8	12 34.5	9 50.0	6 60.3	2 63.8	7 75.9	5 84.5	3 89.7	**2** **93.1**	2 96.6	2 100.0				0.12	8
Cefepime	2 3.4	1 5.2	0 5.2	1 6.9	2 10.3	1 12.1	7 24.1	3 29.3	9 44.8	10 62.1	8 75.9	2 79.3	3 84.5	**9** **100.0**	16	>128
Meropenem	12 37.9	8 51.7	1 53.4	1 55.2	2 58.6	2 62.1	0 62.1	1 63.8	3 69.0					**18** **100.0**	0.06	>8
Piperacillin-tazobactam				0 0.0	1 1.7	3 6.9	7 19.0	3 24.1	3 29.3	5 37.9	1 39.7	4 46.6	3 51.7	**28** **100.0**	128	>128

[a] MIC value is greater than the highest concentration tested.
[b] Comprised of 20 *Escherichia coli*, 36 *Klebsiella pneumoniae*, and 2 *Proteus mirabilis* isolates.

Table 2 Cumulative percent inhibition results for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and comparators against 518 recent *Enterobacteriaceae* isolates (Tier 2)

Organism group (no. tested)/antimicrobials	No. of isolates at MIC (µg/mL; cumulative %) 0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	>[a]	MIC_{50}	MIC_{90}
Enterobacteriaceae (518)[b]																	
Cefepime-AAI101 (fixed 4 µg/mL)	67 14.1	208 54.2	126 78.6	50 88.2	**17** **91.5**	18 95.0	12 97.3	3 97.9	3 98.5	1 98.6	3 99.2	2 99.6	0 99.6	1 99.8	1 100.0	0.03	0.25
Cefepime-AAI101 (fixed 8 µg/mL)	71 13.9	197 51.9	150 80.9	42 89.0	**14** **91.7**	22 95.9	9 97.7	3 98.3	1 98.5	2 98.8	2 99.2	2 99.6	0 99.6	2 100.0		0.03	0.25
Cefepime	62 12.4	180 47.1	124 71.0	35 77.8	22 82.0	16 85.1	14 87.8	9 89.6	**3** **90.2**	13 92.7	9 94.4	12 96.7	3 97.3	1 97.5	13 100.0	0.06	4
Meropenem	194 38.6	166 70.7	**103** **90.5**	26 95.6	5 96.5	3 97.1	3 97.7	2 98.1	1 98.3	3 98.8					**6** **100.0**	0.03	0.06
Piperacillin-tazobactam			0 0.0	2 0.4	39 7.9	38 15.3	83 31.3	179 65.8	76 80.5	27 85.7	22 90.0	**7** **91.3**	12 93.6	12 95.9	21 100.0	2	32

[a] MIC value is greater than the highest concentration tested.
[b] Comprised of 21 *Citrobacter freundii* species complex, 21 *C. koseri*, 28 *Enterobacter aerogenes*, 77 *E. cloacae* species complex, 103 *Escherichia coli*, 27 *Klebsiella oxytoca*, 101 *K. pneumoniae*, 27 *Morganella morganii*, 25 *Proteus mirabilis*, 21 *P. vulgaris* group, 21 *Providencia rettgeri*, 21 *P. stuartii*, and 25 *Serratia marcescens*. There is no redundancy between the clinical isolates used in Tier 1 and those used in Tier 2.

Results

- MIC values for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL), cefepime, meropenem, piperacillin-tazobactam, and/or AAI101 were determined for the Tier 1 (Table 1) and Tier 2 (Table 2) panels of *Enterobacteriaceae*, which included ESBL-phenotype and carbapenem-resistant clinical isolates
- Disk zone diameters were determined for the Tier 2 collection using cefepime-AAI101 30/20 µg and cefepime-AAI101 30/10 µg disks
- Applying CLSI error-rate bounded analysis and CLSI breakpoint interpretive criteria (Figures 2-3) to Tier 2 data, cefepime-AAI101 30/20 µg disks delivered acceptable error rates with no very major errors, no major errors, and a minor error rate (I+1 to I-1) of <40% [20.0%-25.0%] for cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) and for cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL)
 - Proposed disk diffusion breakpoints of ≥25 mm (susceptible) and ≤19 mm (resistant) were calculated for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) combinations
- Applying CLSI error-rate bounded analysis and EUCAST breakpoint interpretive criteria (Figures 4-5) to Tier 2 data, cefepime-AAI101 30/20 µg disks delivered acceptable error rates with no very major errors, 1 major error [*Serratia marcescens*], and a minor error rate (I+1 to I-1) of <40% [18.4%-23.3%] for both cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) and cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL)
 - Proposed disk diffusion breakpoints of ≥25 mm (susceptible) and ≤20 mm (resistant) were calculated for both cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) combinations

Figure 4 Proposed disk breakpoints based on the error-rate bounded method for cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) MIC *vs.* cefepime-AAI101 30/20 µg disks (EUCAST cefepime MIC breakpoint criteria for *Enterobacteriaceae* applied)

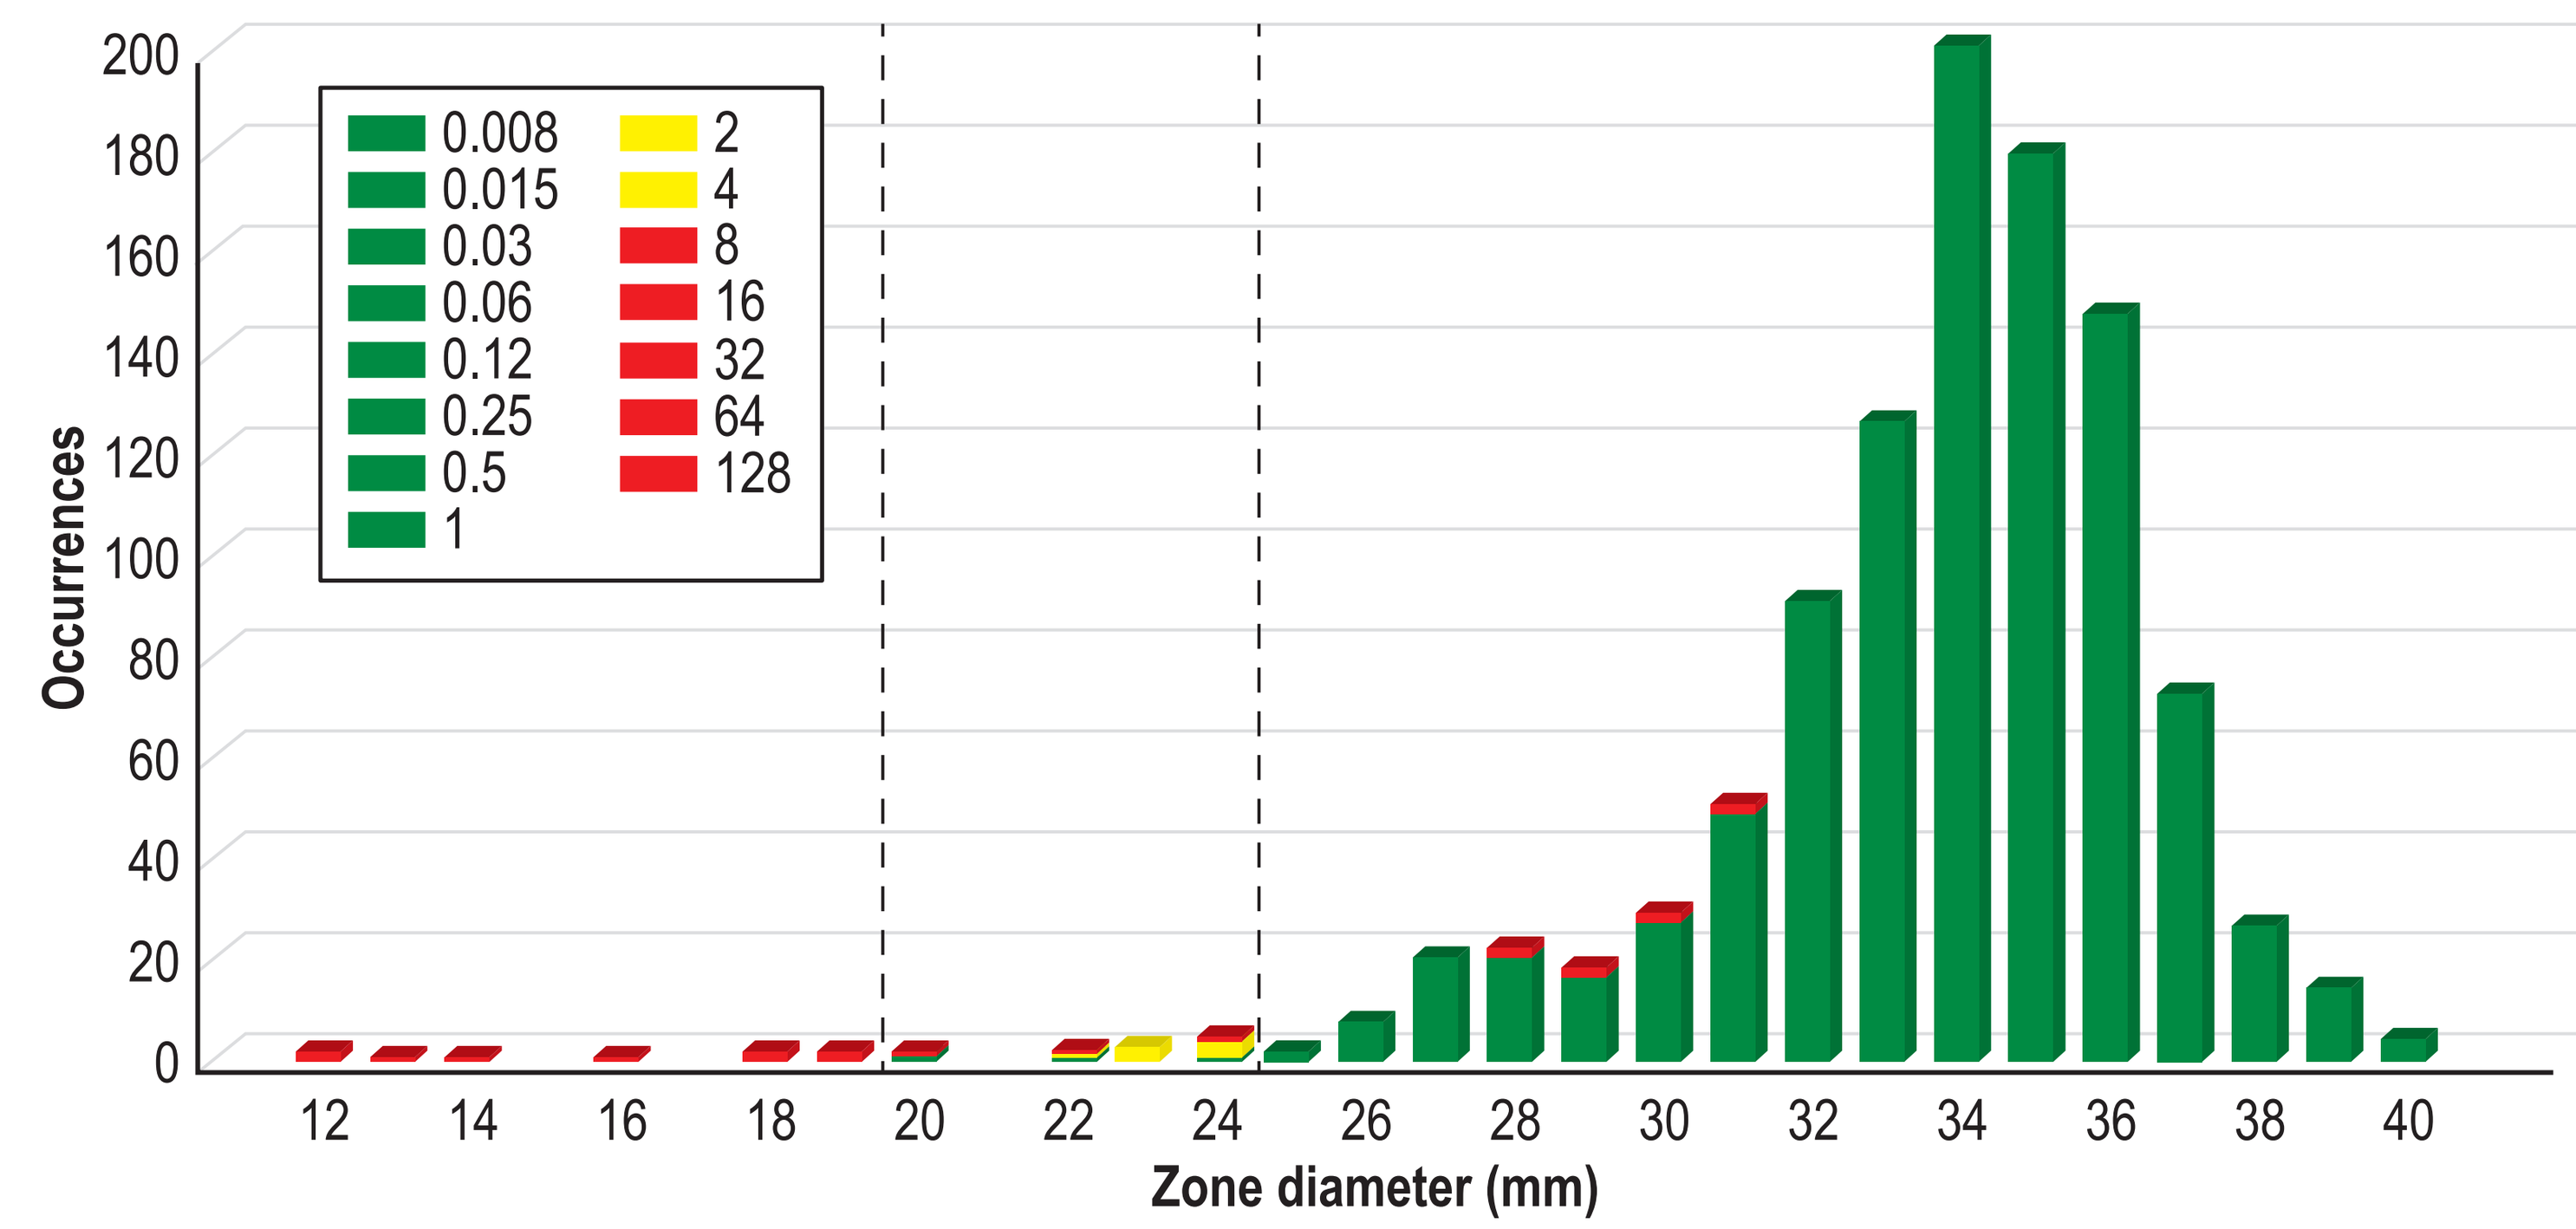
Occurrences
200
180
160
140
120
100
80
60
40
20
0
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
128
12
14
16
18
20
22
24
26
28
30
32
34
36
38
40
Zone diameter (mm)

EUCAST MIC breakpoint criteria for cefepime of ≤1 mg/L (susceptible) and >4 mg/L (resistant) and zone diameter breakpoint criteria of ≥25 mm (susceptible) and ≤19 mm (resistant) applied to cefepime-AAI101 for data analysis purposes

Figure 5 Proposed disk breakpoints based on the error-rate bounded method for cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL) MIC *vs.* cefepime-AAI101 30/20 µg disks (EUCAST cefepime MIC breakpoint criteria for *Enterobacteriaceae* applied)

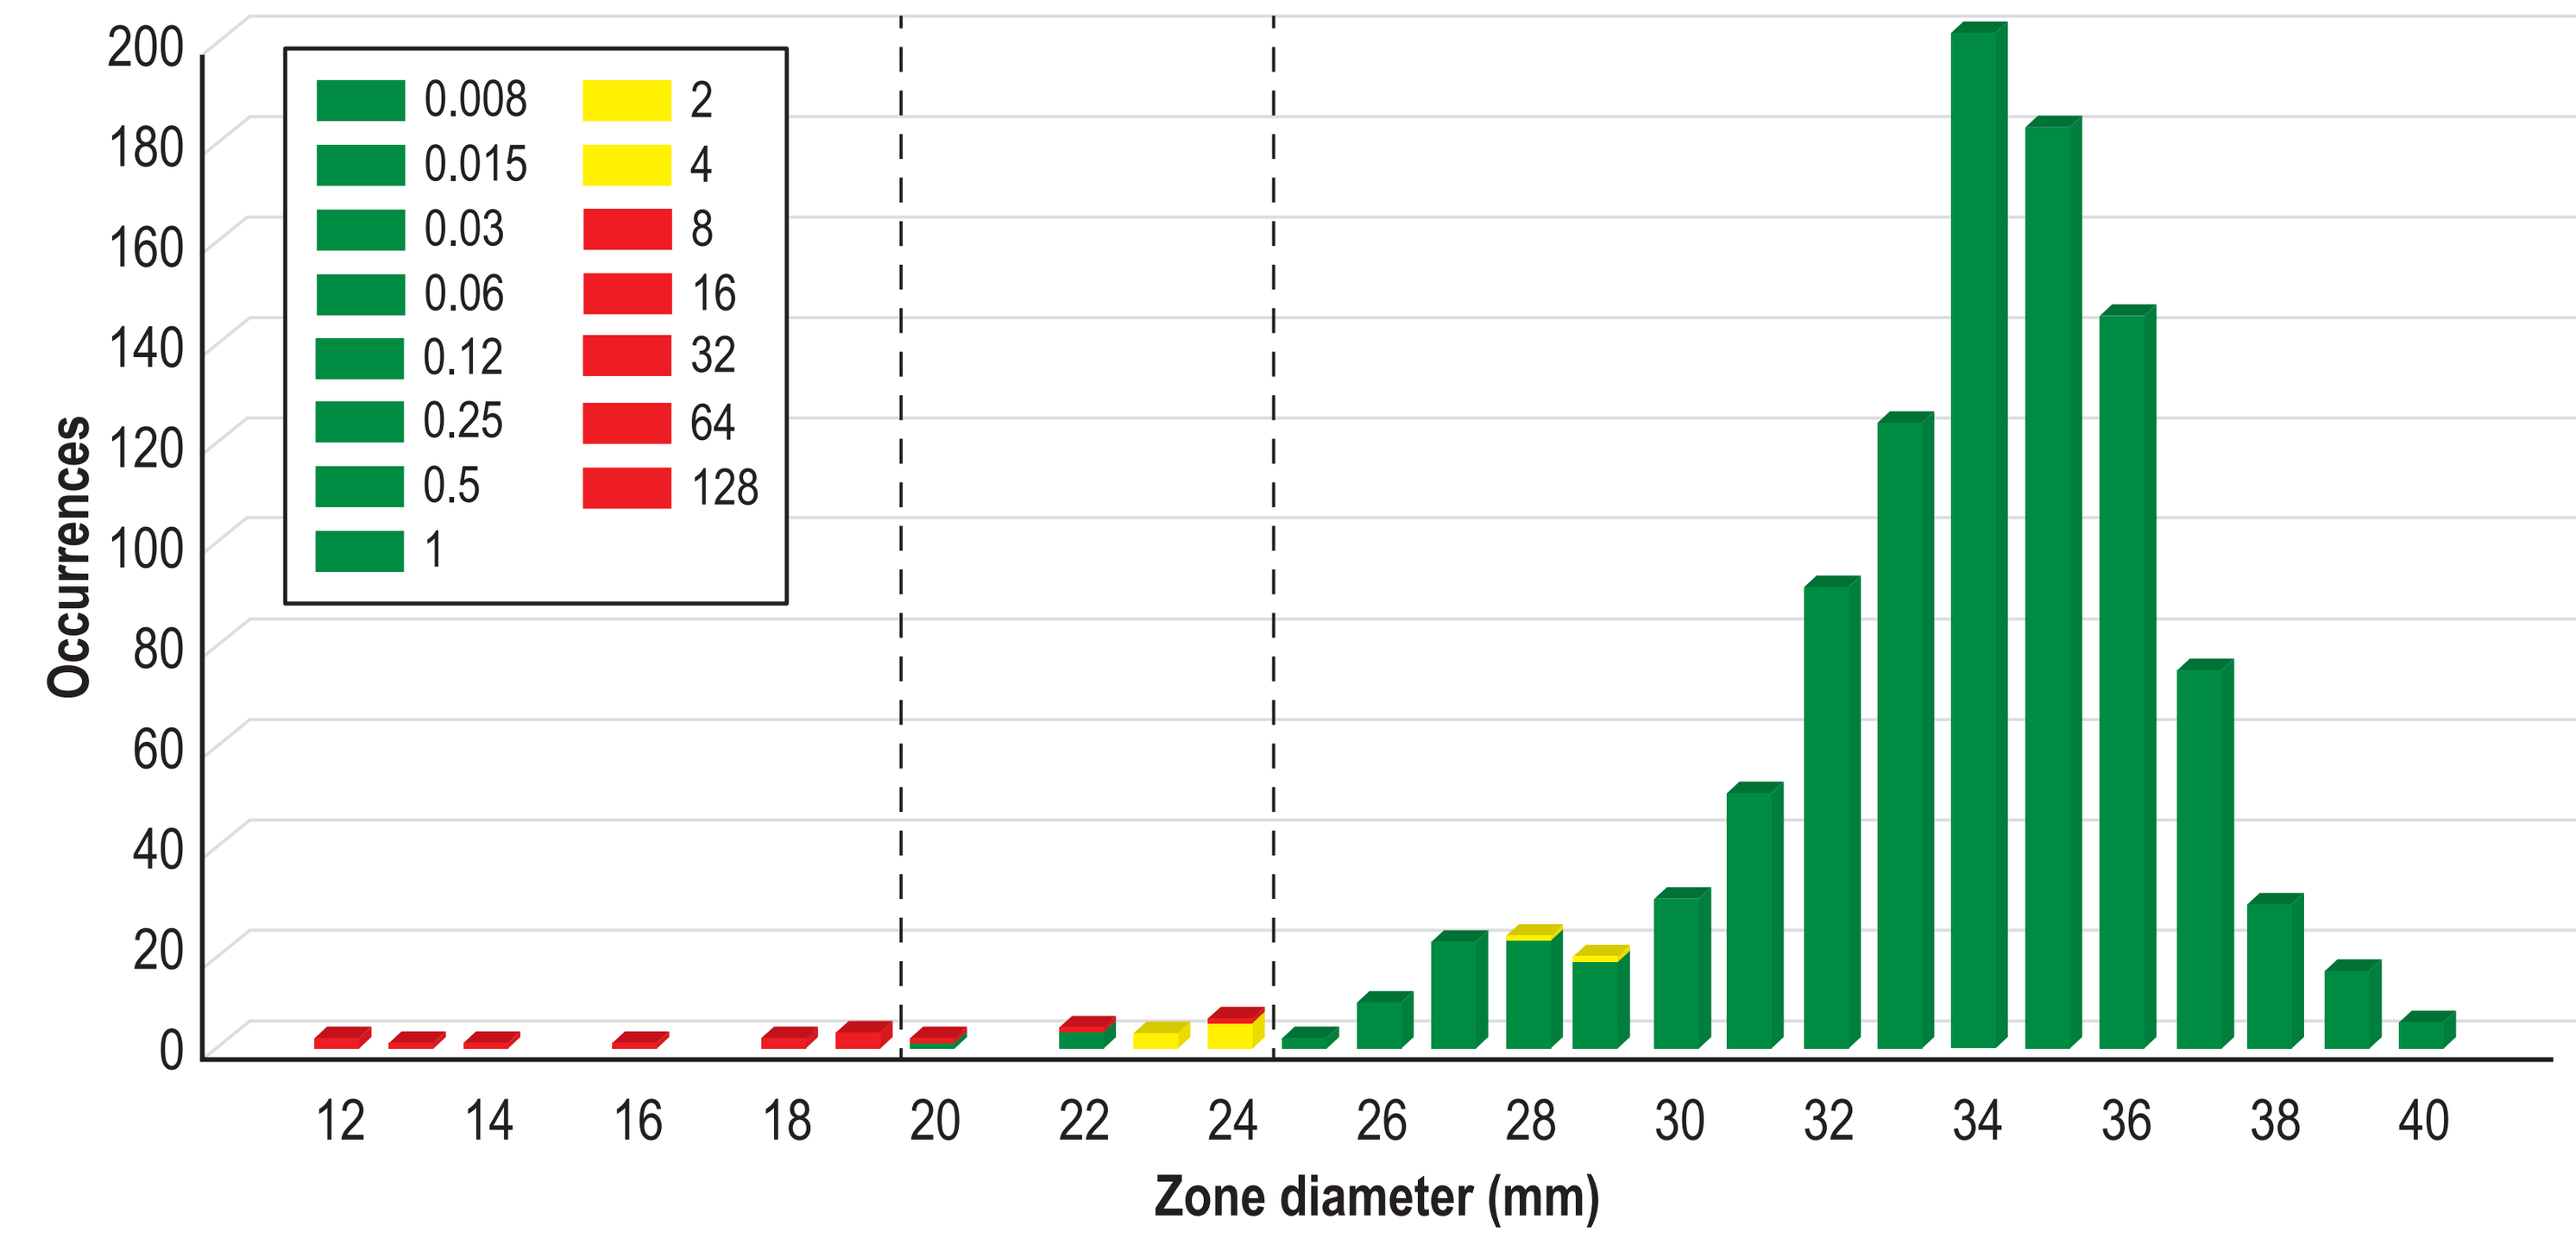
Occurrences
200
180
160
140
120
100
80
60
40
20
0
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
128
12
14
16
18
20
22
24
26
28
30
32
34
36
38
40
Zone diameter (mm)

EUCAST MIC breakpoint criteria for cefepime of ≤1 mg/L (susceptible) and >4 mg/L (resistant) and zone diameter breakpoint criteria of ≥25 mm (susceptible) and ≤19 mm (resistant) applied to cefepime-AAI101 for data analysis purposes

Conclusions

- Proposed CLSI interpretive criteria for cefepime-AAI101 30/20 µg disks compared to cefepime-AAI101 MIC values (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) met CLSI criteria for
 - a cefepime disk concentration that conforms to the concentration accepted for "cefepime-only" disks by the CLSI (as well as by EUCAST);
 - susceptible isolates in the inhibition zone range of 15-35 mm;
 - adequate discrimination between targeted susceptible and resistant populations;
 - acceptable very major, major, and minor error rates; and
 - reproducibility using a minimum number of replicates/ isolates (n = 2)
- Proposed CLSI interpretive criteria for cefepime-AAI101 30/10 µg disks met CLSI criteria when compared to cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) MIC values

Acknowledgements

This study and poster presentation were funded by a grant from Allecra Therapeutics SAS (Saint-Louis, France).

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—tenth edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2016). *M23-A4. Development of in vitro susceptibility testing criteria and quality control parameters: fourth edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2015). *M02-A12. Performance standards for antimicrobial disk susceptibility tests; approved standard—twelfth edition.* Wayne, PA: CLSI.

EUCAST (2018). Breakpoint tables for interpretation of MICs and zone diameters. Version 8.0, January 2018. Available at http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_8.0_Breakpoint_Tables.pdf. Accessed January 2018.