ECCMID 2018

Poster #P2492

Cefepime-AAI101 and Cefepime MIC Quality Control Ranges Using a CLSI M23-A5 Multi-Laboratory Study Design

MD Huband[1], S Shapiro[2], KA Fedler[1], RK Flamm[1]

[1]JMI Laboratories, North Liberty, Iowa, USA; [2]Allecra Therapeutics, Saint-Louis, France

Contact Information:
Michael D. Huband, Associate Director
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: michael-huband@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit http://www.jmilabs.com/data/posters/ECCMID2018-cefepime-AAI101-M23.pdf

Charges may apply.
No personal information is stored.

Introduction

- AAI101 is a novel β-lactamase inhibitor highly active against extended-spectrum β-lactamases (ESBLs), the primary mechanism of β-lactam resistance toward oxyiminocephalosporins
- The cefepime-AAI101 combination has completed Phase 2 clinical trials to evaluate the efficacy and safety of the combination cefepime-AAI101 for treatment of complicated urinary tract infections (cUTI)
- Clinical and Laboratory Standards Institute (CLSI) M23-A4 (tier 2) quality control studies were conducted to establish cefepime-AAI101 broth microdilution (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and cefepime quality control (QC) ranges against CLSI QC reference strains

Materials and Methods

- Investigators and institutions participating in CLSI M23 (2018) broth microdilution QC studies for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and cefepime are listed in Table 1
- Broth microdilution studies utilized a minimum of 7 participating laboratories (Table 1), 3 lots of cation-adjusted Mueller-Hinton medium obtained from at least 2 different manufacturers, and ≥10 replicate tests per QC strain tested over a minimum of 3 days
 - Cation adjusted Mueller-Hinton medium for broth microdilution testing was obtained from Difco (Detroit, Michigan), Becton Dickinson (BD; Sparks, Maryland), and Oxoid (Hampshire, United Kingdom)
- Frozen-form broth microdilution susceptibility panels were prepared in a certified good-manufacturing practice facility (Trek Diagnostic Systems/Thermo Fisher Scientific, Oakwood Village, Ohio) using AAI101 powder provided by Allecra Therapeutics
- QC reference strains tested included *Escherichia coli* ATCC 25922, *E. coli* ATCC 35218, *E. coli* NCTC 13353, *Klebsiella pneumoniae* ATCC 700603, and *Pseudomonas aeruginosa* ATCC 27853
- *Escherichia coli* NCTC 13353 was included as an ESBL-producing (CTX-M-15) QC reference strain specifically to address the activity of the cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) combinations

Table 1 Investigators and laboratories participating in the cefepime-AAI101 and cefepime M23 broth microdilution quality control studies

Investigator	Laboratory and location	M23 study participation[a]
R. Flamm, PhD	JMI Laboratories, North Liberty, Iowa, USA	F4 and F8
C. Knapp, MS	Thermo Fisher Scientific, Oakwood Village, Ohio, USA	F4 and F8
J. Hindler, MS	UCLA Medical Center, Los Angeles, California, USA	F4 and F8
G. Kallstrom, PhD	Summa Health System, Akron, Ohio, USA	F8
E. Munson, PhD	Wheaton Franciscan Laboratory, Wauwatosa, Wisconsin, USA	F8
S. Riedel, MD, PhD	Beth Israel Deaconess Medical Center, Boston, Massachusetts, USA	F4 and F8
D. Hardy, PhD	University of Rochester Medical Center, Rochester, New York, USA	F4 and F8
T. Fritsche, MD, PhD	Marshfield Laboratories, Marshfield, Wisconsin, USA	F4 and F8
C. Pillar, PhD	Micromyx Inc., Kalamazoo, Michigan, USA	F4
G. Denys, PhD	Indiana University Health, Methodist Hospital, Indianapolis, Indiana, USA	F4

[a] F4 = fixed AAI101 concentration of 4 µg/mL, F8 = fixed AAI101 concentration of 8 µg/mL

Results

- Applying CLSI M23 and RangeFinder statistical analysis criteria to the cefepime-AAI101 combinations, ≥99.2% of cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) and ≥99.6% of cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL) MIC values from the participating laboratories were within the approved QC ranges for each of the reference strains tested (Table 2)
- Colony counts were performed on each of the QC reference strains tested (Table 3) and results were within acceptable inoculum targets
- Three dilution broth microdilution QC ranges were approved by the CLSI for cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) against: *E. coli* ATCC 25922, *E. coli* ATCC 35218, *E. coli* NCTC 13353, and *K. pneumoniae* ATCC 700603 (Table 2, Table 4, and Figure 1), and a 4-dilution QC range was approved for *P. aeruginosa* ATCC 27853 based on the bimodal (65.7% MIC shoulder at 2/4 µg/mL) distribution of data (Table 2)
- Similarly, 3 dilution broth microdilution QC ranges were approved for cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL) against *E. coli* ATCC 25922, *E. coli* NCTC 13353, *K. pneumoniae* ATCC 700603, and *P. aeruginosa* ATCC 27853 (Table 2, Table 5, and Figure 2), and a 4-dilution QC range was approved for *E. coli* ATCC 35218 based on the bimodal (80.0% MIC shoulder at 0.03/8 µg/mL) distribution of data (Table 2)
- Among the reference bacterial strains tested that have approved cefepime QC ranges, 100.0% of the cefepime MIC values against *E. coli* ATCC 25922, *E. coli* ATCC 35218, *E. coli* NCTC 13353, and *P. aeruginosa* ATCC 27853 and 99.8% of the cefepime MIC results against *K. pneumoniae* ATCC 700603 were within CLSI approved QC ranges, providing validated internal controls for these studies

Table 2 CLSI approved broth microdilution quality control ranges for cefepime-AAI101 and cefepime against reference strains

Quality control reference strain	CLSI approved quality control ranges (µg/mL) (# of dilutions; % of values in range)		
	Cefepime-AAI101 (fixed 4 µg/mL)	Cefepime-AAI101 (fixed 8 µg/mL)	Cefepime
Escherichia coli ATCC 25922	0.03/4 – 0.12/4 (3; 100.0%)	0.03/8 – 0.12/8 (3; 100.0%)	0.016 – 0.12[a] (4; 100.0%)
E. coli ATCC 35218	0.016/4 – 0.06/4 (3; 100.0%)	0.008/8 – 0.06/8 (4; 100.0%)	0.008 – 0.06[b] (4; 100.0%)
E. coli NCTC 13353	0.06/4 – 0.25/4 (3; 99.5%)	0.03/8 – 0.12/8 (3; 100.0%)	≥64[b] (100.0%)
Klebsiella pneumoniae ATCC 700603	0.12/4 – 0.5/4 (3; 99.2%)	0.12/8 – 0.5/8 (3; 100.0%)	0.5 – 2[a] (3; 99.8%)
Pseudomonas aeruginosa ATCC 27853	0.5/4 – 4/4 (4; 100.0%)	0.5/8 – 2/8 (3; 99.6%)	0.5 – 4[a] (4; 100.0%)

[a] Current CLSI quality control range
[b] QC range approved at the June 2017 CLSI meeting (Philadelphia, Pennsylvania)

Table 3 Colony counts for the quality control reference strains used in broth microdilution susceptibility testing

CLSI QC reference strain	Range[a] (CFU/mL)	Average[b] (CFU/mL)
Escherichia coli ATCC 25922	8.0 x 10^4 – 9.0 x 10^5	3.1 x 10^5
E. coli ATCC 35218	1.0 x 10^5 – 8.0 x 10^5	3.1 x 10^5
E. coli NCTC 13353	1.0 x 10^5 – 7.1 x 10^5	3.2 x 10^5
Klebsiella pneumoniae ATCC 700603	1.0 x 10^5 – 9.0 x 10^5	3.2 x 10^5
Pseudomonas aeruginosa ATCC 27853	9.0 x 10^4 – 9.5 x 10^5	4.0 x 10^5

[a] Range of all participating laboratories
[b] Average of all participating laboratories

Table 4 Inter- and intralaboratory comparisons of cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) MIC values against *E. coli* NCTC 13353 for an 8-laboratory protocol meeting the study design guidelines found in CLSI M23-A4 (2016)

MIC (µg/mL)	Occurrences by media lot[a]			Laboratory (occurrences):								Total
	A	B	C	A	B	C	D	E	F	G	H[b]	
0.03/4												
0.06/4	10	11	8	1		2	1		20	5		29
0.12/4	51	53	50	12	25	27	25	30	10	25		154
0.25/4	8	6	12	16	5	1	4					26
0.5/4	1			1							19	1
1/4											10	
2/4											1	
Total	70	70	70	30	30	30	30	30	30	30	—	210
Mean	0.13/4	0.12/4	0.14/4	0.20/4	0.14/4	0.12/4	0.14/4	0.12/4	0.08/4	0.11/4	—	0.13/4
Median	0.12/4	0.12/4	0.12/4	0.25/4	0.12/4	0.12/4	0.12/4	0.12/4	0.06/4	0.12/4	—	0.12/4
Mode	0.12/4	0.12/4	0.12/4	0.25/4	0.12/4	0.12/4	0.12/4	0.12/4	0.06/4	0.12/4	—	0.12/4
Geometric mean	0.12/4	0.11/4	0.13/4	0.18/4	0.14/4	0.12/4	0.13/4	0.12/4	0.08/4	0.11/4		0.12/4
Range	4	3	3	4	2	3	3	1	2	2	—	4

[a] A, Difco lot # 5181782; B, BD lot # 5257869; C, Oxoid lot # 1433705
[b] Laboratory H is a statistical outlier for the mean, median, and modal MIC values and was excluded from the data analysis

Approved QC range (µg/mL)	# of dilutions	% in range
0.06/4 – 0.25/4	3	99.5% (209/210)

Figure 1 Cefepime-AAI101 (fixed AAI101 concentration of 4 µg/mL) MIC distributions by medium lot for *Escherichia coli* NCTC 13353

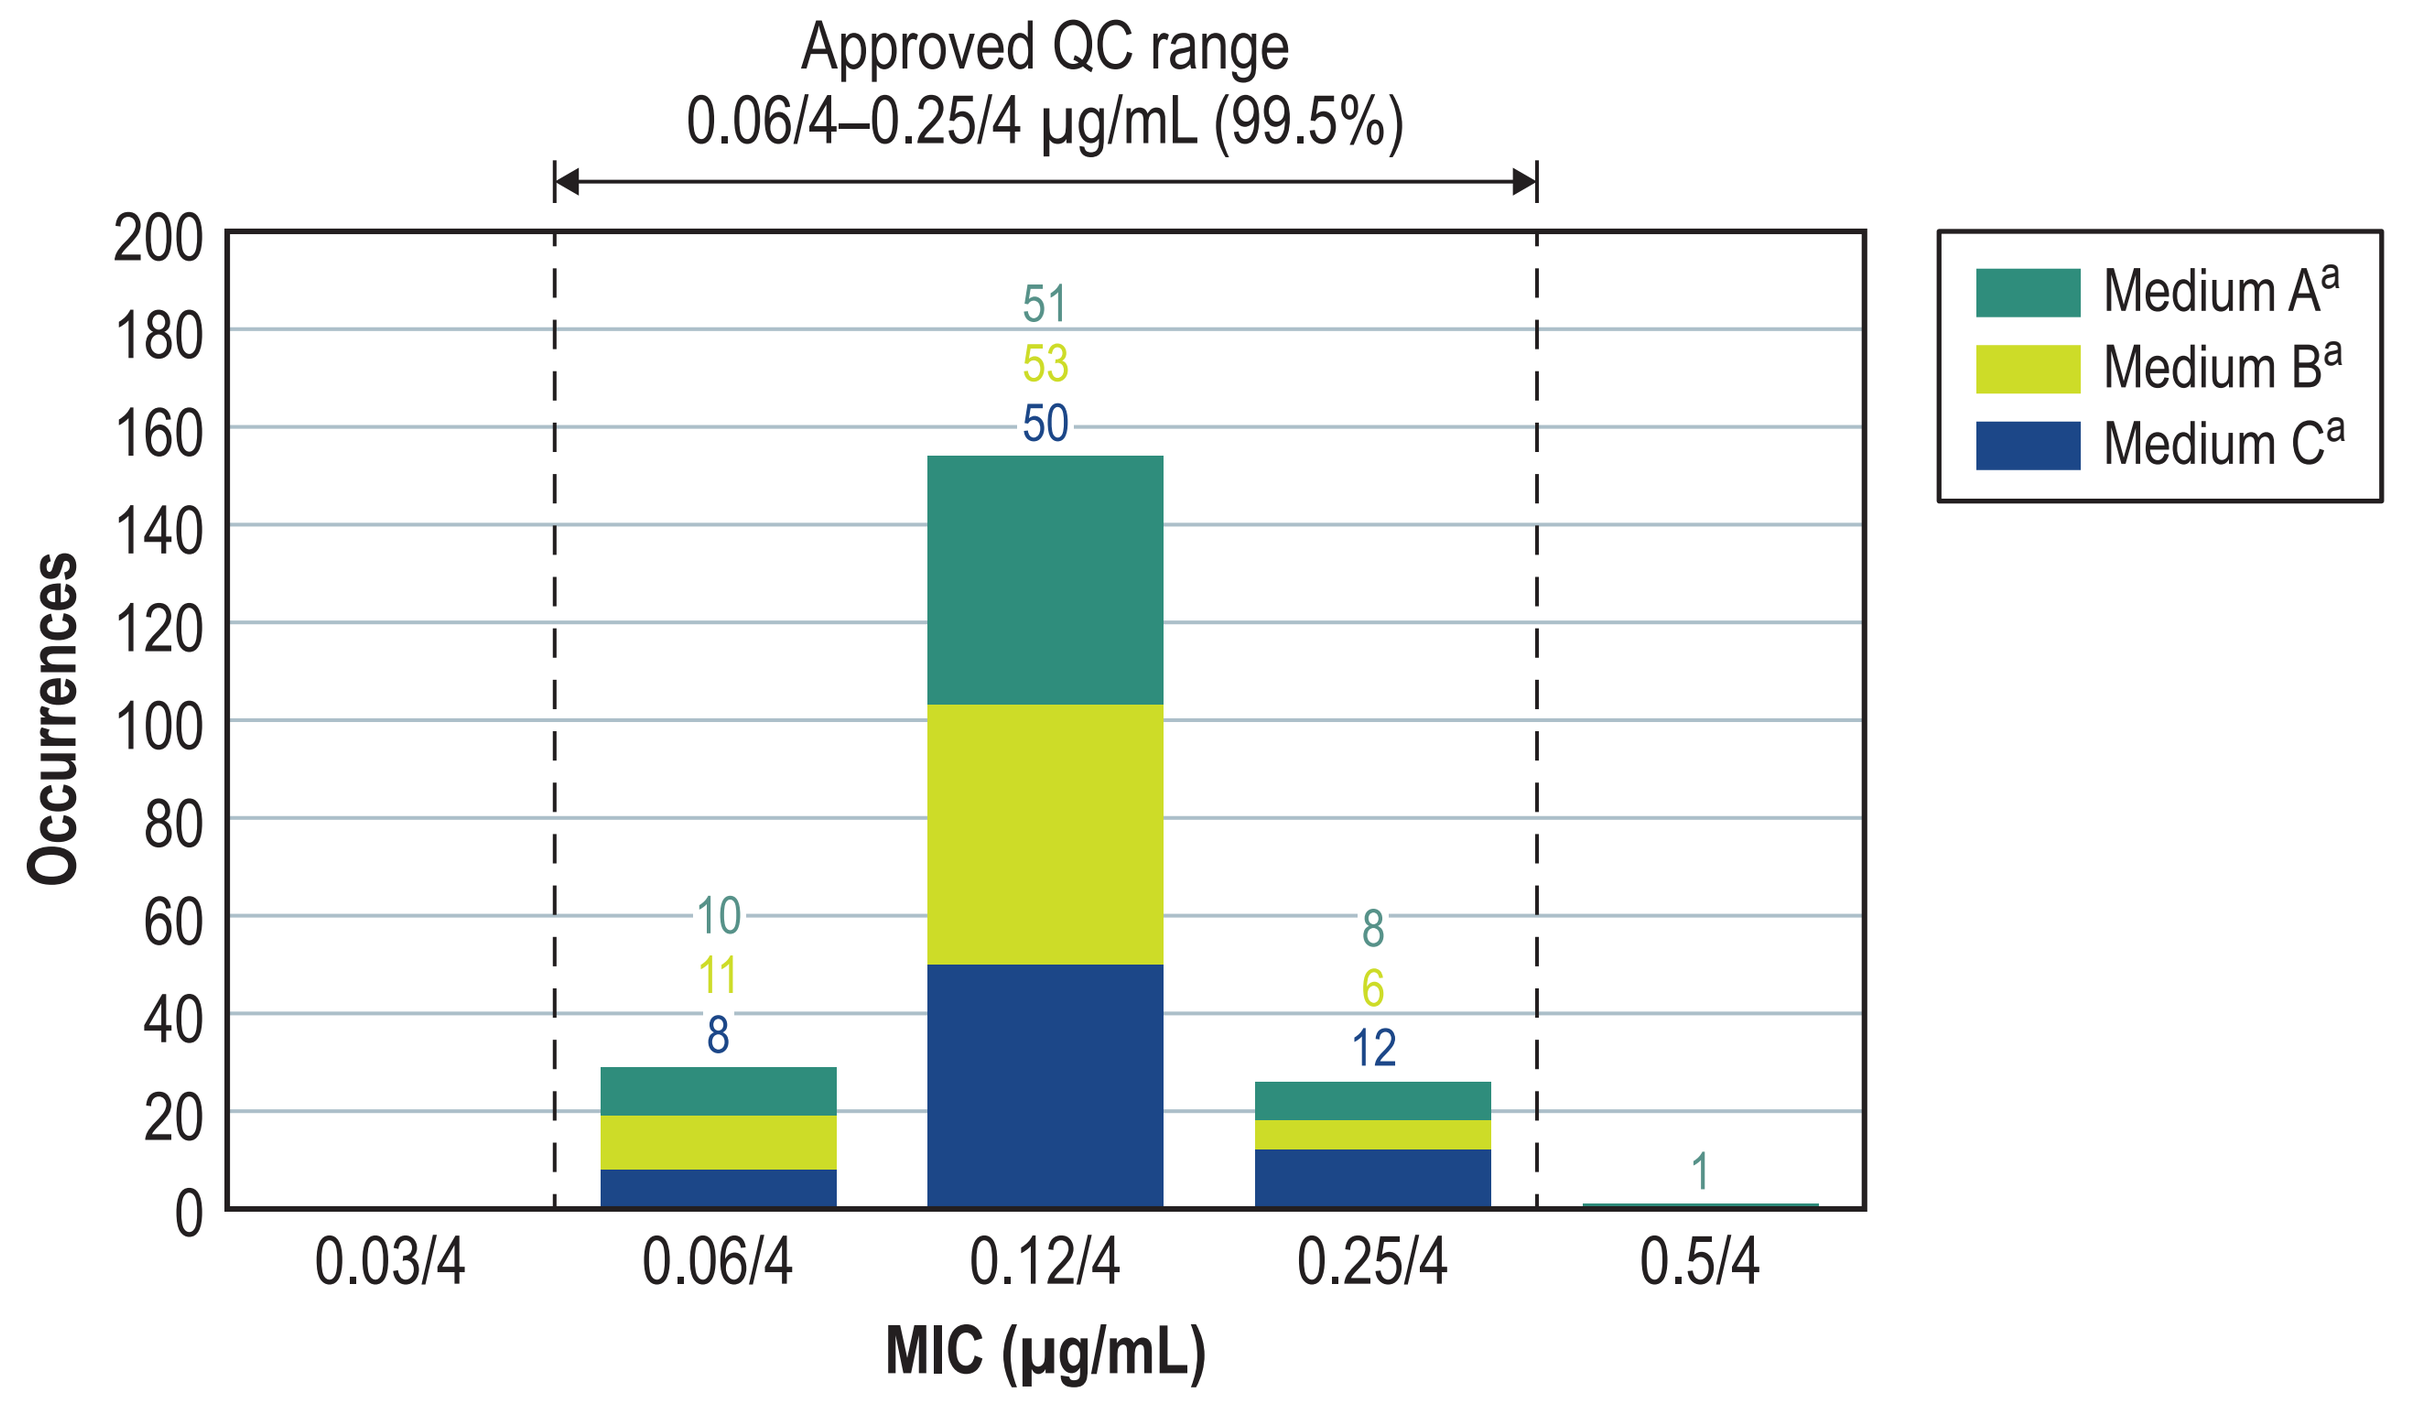

Approved QC range
0.06/4–0.25/4 µg/mL (99.5%)
Occurrences
200
180
160
140
120
100
80
60
40
20
0
10
11
8
51
53
50
8
6
12
1
0.03/4
0.06/4
0.12/4
0.25/4
0.5/4
MIC (µg/mL)
Medium A[a]
Medium B[a]
Medium C[a]

[a] Medium A, Difco (lot 5181782); Medium B, BD (lot 5257869); Medium C, Oxoid (lot 1433705)

Figure 2 Cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL) MIC distributions by medium lot for *Escherichia coli* NCTC 13353

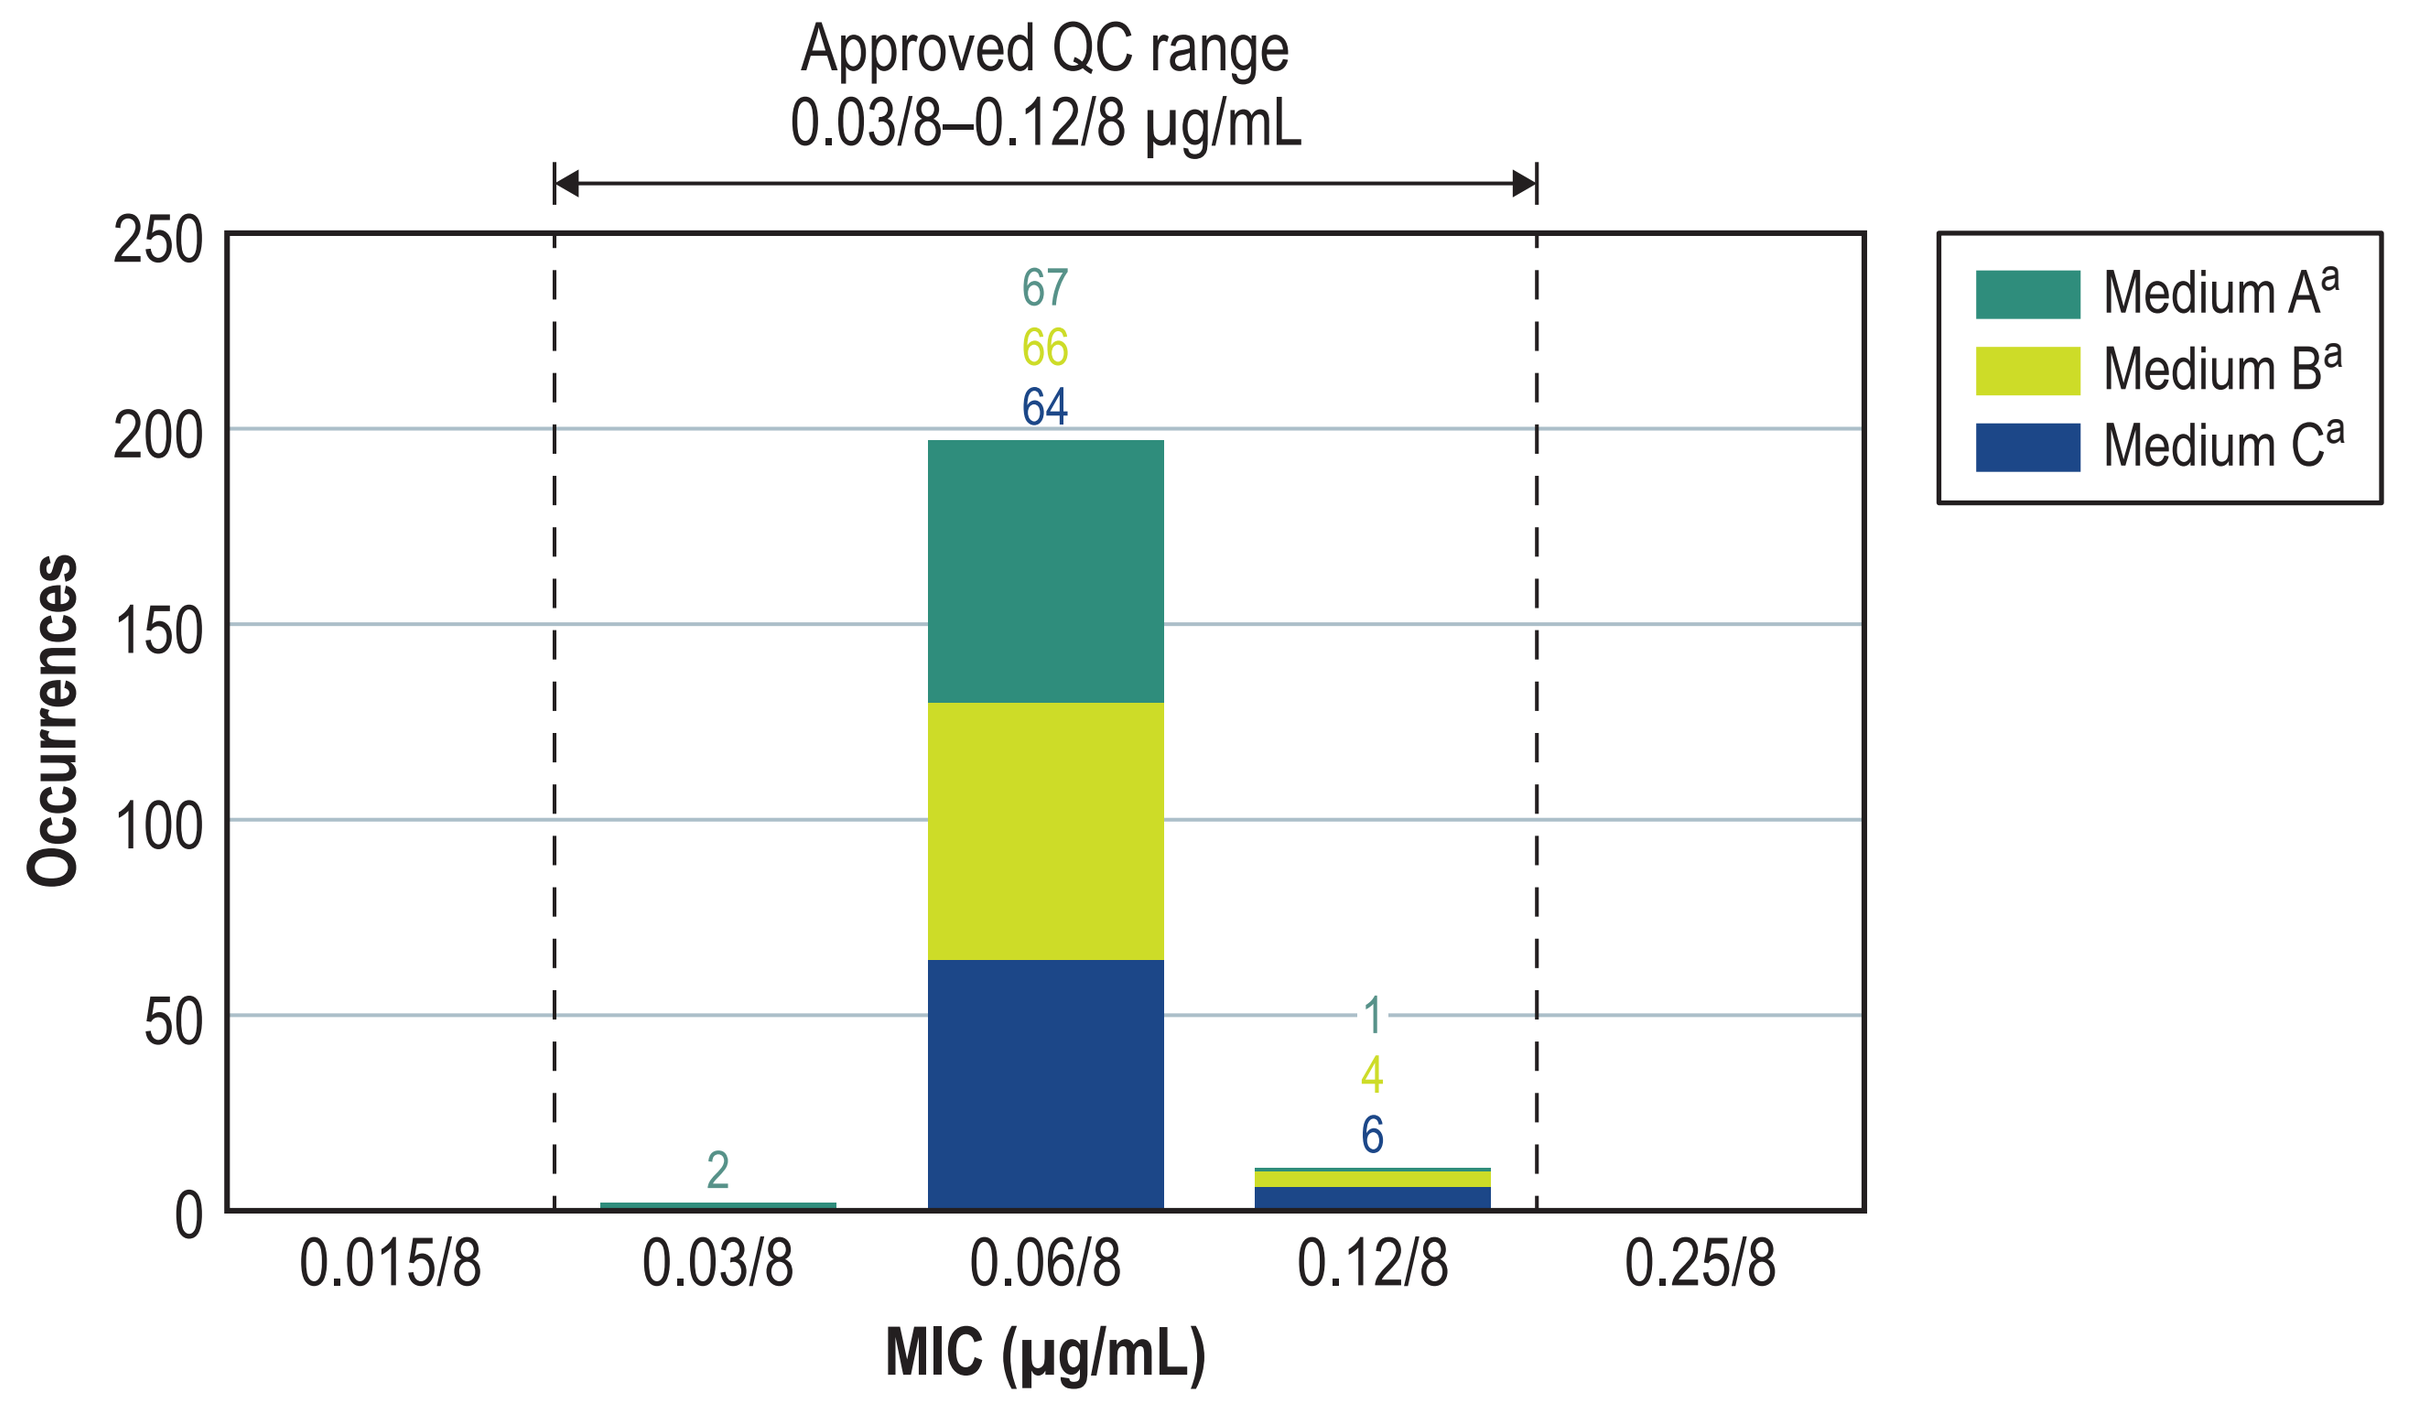

Approved QC range
0.03/8–0.12/8 µg/mL
Occurrences
250
200
150
100
50
0
2
67
66
64
1
4
6
0.015/8
0.03/8
0.06/8
0.12/8
0.25/8
MIC (µg/mL)
Medium A[a]
Medium B[a]
Medium C[a]

[a] Medium A, Difco (lot 5181782); Medium B, BD (lot 5257869); Medium C, Oxoid (lot 1433705)

Table 5 Inter- and intralaboratory comparisons of cefepime-AAI101 (fixed AAI101 concentration of 8 µg/mL) MIC values against *E. coli* NCTC 13353 for an 8-laboratory protocol meeting the study design guidelines found in CLSI M23-A4 (2016)

MIC (µg/mL)	Occurrences by media lot[a]			Laboratory (occurrences):								Total
	A	B	C	A	B	C	D	E	F[b]	G	H	
0.015/8												
0.03/8	2			1		1						2
0.06/8	67	66	64	29	24	29	29	30	1	28	28	197
0.12/8	1	4	6		6		1			2	2	11
0.25/8									18			
0.5/8									11			
Total	70	70	70	30	30	30	30	30	30	30	30	210
Mean	0.06/8	0.06/8	0.07/8	0.06/8	0.07/8	0.06/8	0.06/8	0.06/8	0.34/8	0.07/8	0.07/8	0.06/8
Median	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.25/8	0.06/8	0.06/8	0.06/8
Mode	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.06/8	0.25/8	0.06/8	0.06/8	0.06/8
Geometric mean	0.06/8	0.06/8	0.06/8	0.06/8	0.07/8	0.06/8	0.06/8	0.06/8	0.31/8	0.06/8	0.06/8	0.06/8
Range	3	2	2	2	2	2	2	1	4	2	2	3

[a] A, Difco lot # 5181782; B, BD lot # 5257869; C = Oxoid lot # 1433705
[b] Laboratory F is an outlier for the mean, median, and modal MIC values and was excluded from the data analysis

Approved QC range (µg/mL)	# of dilutions	% in range
0.03/8 – 0.12/8	3	100.0% (210/210)

Conclusions

- Cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) broth microdilution susceptibility testing demonstrated acceptable intra- and interlaboratory reproducibility with the following CLSI QC reference strains: *E. coli* ATCC 25922, *E. coli* ATCC 35218, *E. coli* NCTC 13353, *K. pneumoniae* ATCC 700603, and *P. aeruginosa* ATCC 27853
- At its June 2017 meeting, the CLSI Subcommittee on Antimicrobial Susceptibility Testing (AST) approved cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) broth microdilution QC ranges against the following reference strains: *E. coli* ATCC 25922, *E. coli* ATCC 35218, *E. coli* NCTC 13353, *K. pneumoniae* ATCC 700603, and *P. aeruginosa* ATCC 27853
- At its June 2017 meeting, the CLSI Subcommittee on AST also approved the cefepime broth microdilution QC range for *E. coli* ATCC 35218 and updated the QC range for *E. coli* NCTC 13353
- Overall, 99.2%-100.0% of cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and 99.8%-100.0% of cefepime MIC values against the reference strains tested were within the QC ranges approved by the CLSI
- *E. coli* NCTC 13353 (CTX-M-15; ESBL phenotype), which is resistant to cefepime, provides adequate QC for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL), and should be tested routinely when evaluating the activity of cefepime-AAI101 combinations
- The approved broth microdilution QC ranges for cefepime and for cefepime-AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) will assist clinical and reference laboratories in generating reliable, accurate susceptibility testing results during clinical trials and in clinical microbiology practice

Acknowledgements

This study and abstract presentation were funded by a research grant from Allecra Therapeutics.

References

Clinical and Laboratory Standards Institute (2018). *M23-A5. Development of in vitro susceptibility testing criteria and quality control parameters: fifth edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition*. Wayne, PA: CLSI.

Turnidge J, Bordash G (2007). Statistical methods for establishing quality control ranges for antibacterial agents in Clinical and Laboratory Standards Institute susceptibility testing. *Antimicrob Agents Chemother* 51: 2483–2488.